A Comparison of VAMS™ Technology versus Traditional DBS Sampling in a Ligand Binding Assay

Daniel Burns, Beth Johnson, Jin Wang, Mark Ma

Amgen Inc, Thousand Oaks, CA

ABSTRACT

Dried Blood Spots (DBS) have been used in place of serum and plasma in order to reduce animal use, lower sample volume requirements in pediatric studies, simplify handling, storage, and shipping of samples, and facilitate home based and third world country studies where refrigeration and lab equipment are lacking. However, DBS implementation has been limited by industrial and regulatory concerns particularly of the hematocrit effect on method accuracy. Volumetric Absorption Matrix Sampling (VAMS™), a second generation of DBS technology, has been developed to provide precise sampling for accuracy, simplified sample processing, and to eliminate hematocrit effect. In this poster we evaluated VAMS™ technology by testing a variety of validation parameters which were compared with results obtained with traditional DBS samples using the same ligand binding Assay (LBA).

Standards, quality control samples, and other test samples were prepared by spiking a therapeutic mAb into fresh human whole blood. VAMS™ samples were prepared by absorbing spiked blood samples onto VAMS™, drying samples for 2 to 24 hours at ambient room temperature (ART), storing samples at prescribed testing conditions, and extracting analyte from VAMS™ for sample analysis. The evaluated validation parameters were accuracy and precision, extraction efficiency, extracted matrix effect, selectivity, dilutional linearity, hematocrit effect, and stability.

The method demonstrated a linear range of 10.0–1000 ng/mL with 4-parameter logistic regression. Inter-assay accuracy was ≤ 7.3%, inter-assay precision was ≤ 11.1, and the total error was ≤ 18.4%. An average of 89.5% extraction efficiency was achieved. There was no observed selectivity issue, hematocrit effect, or extracted matrix effect on the assay. The dilutional linearity was demonstrated from 1:15 to 1:1500. One-month stability was demonstrated at ART, 4°C, 37°C, and –0°C storage conditions.

We have successfully evaluated the implementation of VAMS™ technology in LBAs. The validation parameter results revealed comparable assay performance between VAMS™ versus DBS sampling. VAMS™ technology offered better user-friendly sample processing. In addition, VAMS™ technology overcame the hematocrit effect that was observed with the implementation of DBS.

VAMS TECHNOLOGY

Volumetric Absorption Matrix Sampling (VAMS™) is a next generation DBS system. It overcomes the impact of variable hematocrit levels and homogeneity on sample accuracy that is associated with traditional DBS. VAMS™ helps eliminate these impacts by accurately adsorbing 10 µL of whole blood (WB) sample into hydrophilic polymeric tips which are then extracted as intact tips. Samples are obtained by allowing a tip to touch a drop of WB (without submerging) allowing enough time for the tip to wick up 10 µL of WB. This enables VAMS™ to simplify the process by eliminating the need to spot and core samples which is required for DBS sampling.

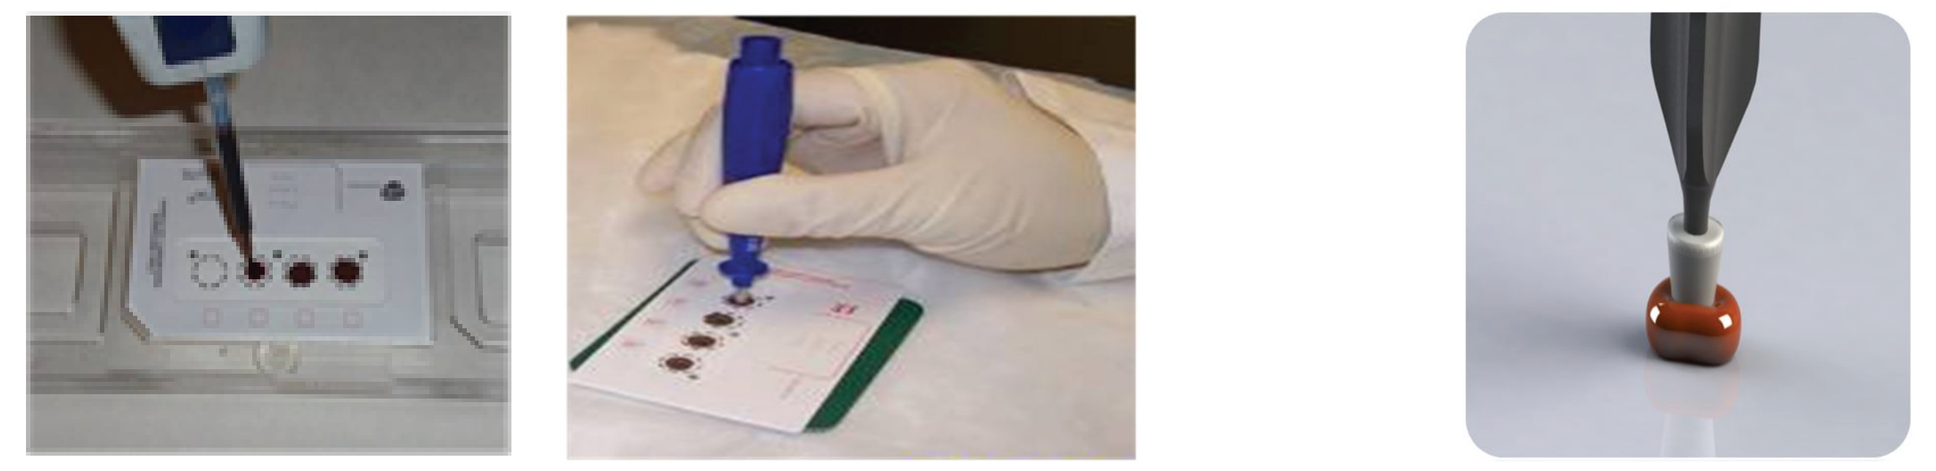

DBS Preparation versus **VAMS™ Preparation**

In this poster we evaluated VAMS™ technology by testing a variety of assay parameters using a Ligand Binding Assay (LBA). We have also compared the VAMS™ evaluation results to the DBS evaluation results that were previously generated using the same LBA.

METHODS

Analytical Method – Assay Format

The biotherapeutic used for the evaluation is a fully human IgG_2 mAb

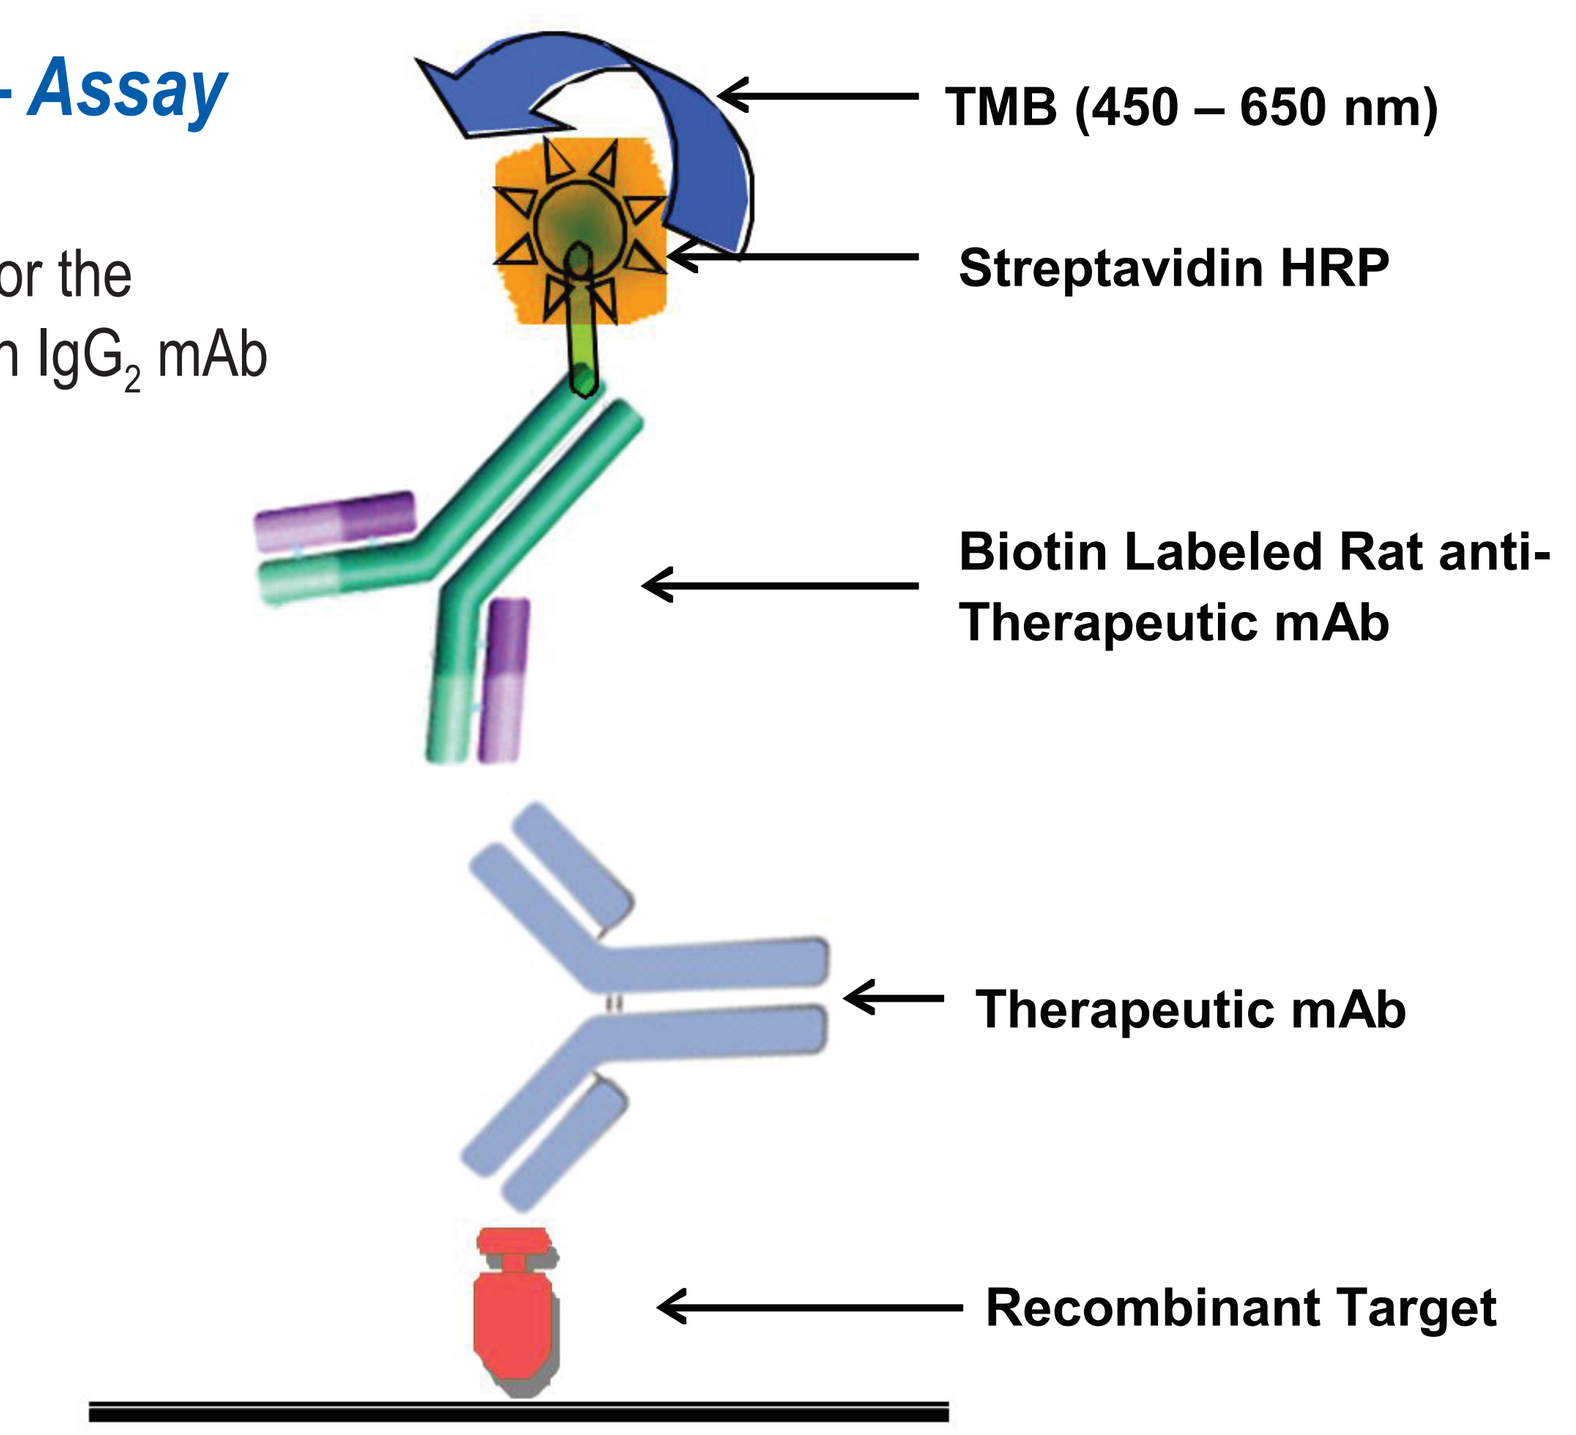

TMB (450 – 650 nm)
Streptavidin HRP
Biotin Labeled Rat anti-Therapeutic mAb
Therapeutic mAb
Recombinant Target

METHODS (Continued)

VAMS™ Preparation

- Step 1 • Draw WB sample and prepare STD/QC by spiking therapeutic mAb into assay buffer at 50x final concentration, dilute 1:50 in WB, and incubate on shaker at 37°C for 1 hr
- Step 2 • WB Sample is absorbed onto a VAMS™ tip and dried for 2 to 24 hours at ART
- Step 3 • Samples not analyzed immediately are stored at ART or various other conditions in racks inserted in zip locked bags with desiccant until a day before analysis
- Step 4 • A day before analysis the VAMS™ tip is placed in a 96 deep well block with extraction buffer
- Step 5 • The extraction block is rotated at 1100 rpm using a platform mixer/rotator for approximately 1 hr and then incubated at 2° to 8°C overnight
- Step 6 • On day of analysis VAMS™ tips are removed, the deep well extraction block is centrifuged, and the supernatant is removed for sample analysis

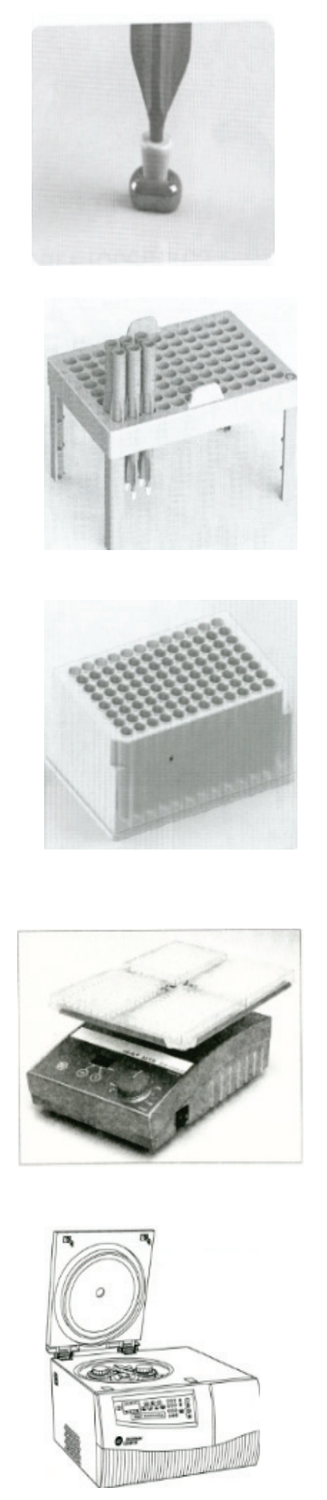

VAMS™ Technology Evaluated Parameters

- Extraction Efficiency
- Extracted Matrix Effect
- Accuracy and Precision
- Selectivity
- Dilutional Linearity
- Hematocrit Effect
- Stability

RESULTS

Extraction Efficiency

A set of samples at LQC and HQC level prepared in WB was absorbed onto VAMS™ and extracted per assay procedures and another set of samples was prepared in extracted blank WB matrix (sample diluent). Both sets were analyzed using the same LBA assay. The extraction efficiency was determined by comparing the % recovery of the extracted samples versus the non-extracted matrix samples.

Table 1: Extraction Efficiency Results

Samples	Extraction Efficiency
LQC	88.4%
HQC	90.6%
Average	89.5%

Extraction Efficiency = Extracted sample result / Matrix sample result X100

Extracted Matrix Effect

A set of samples at LQC and HQC level were prepared in extracted blank WB matrix (sample diluent) and in extraction buffer (Super Block). Both sets were analyzed using the same LBA assay. The % Diff of the samples prepared in blank WB matrix (sample diluent) versus extraction buffer were determined and is displayed in figure 1 below.

RESULTS (Continued)

Figure 1. %Diff of Blank WB Matrix Samples versus Extraction Buffer Samples

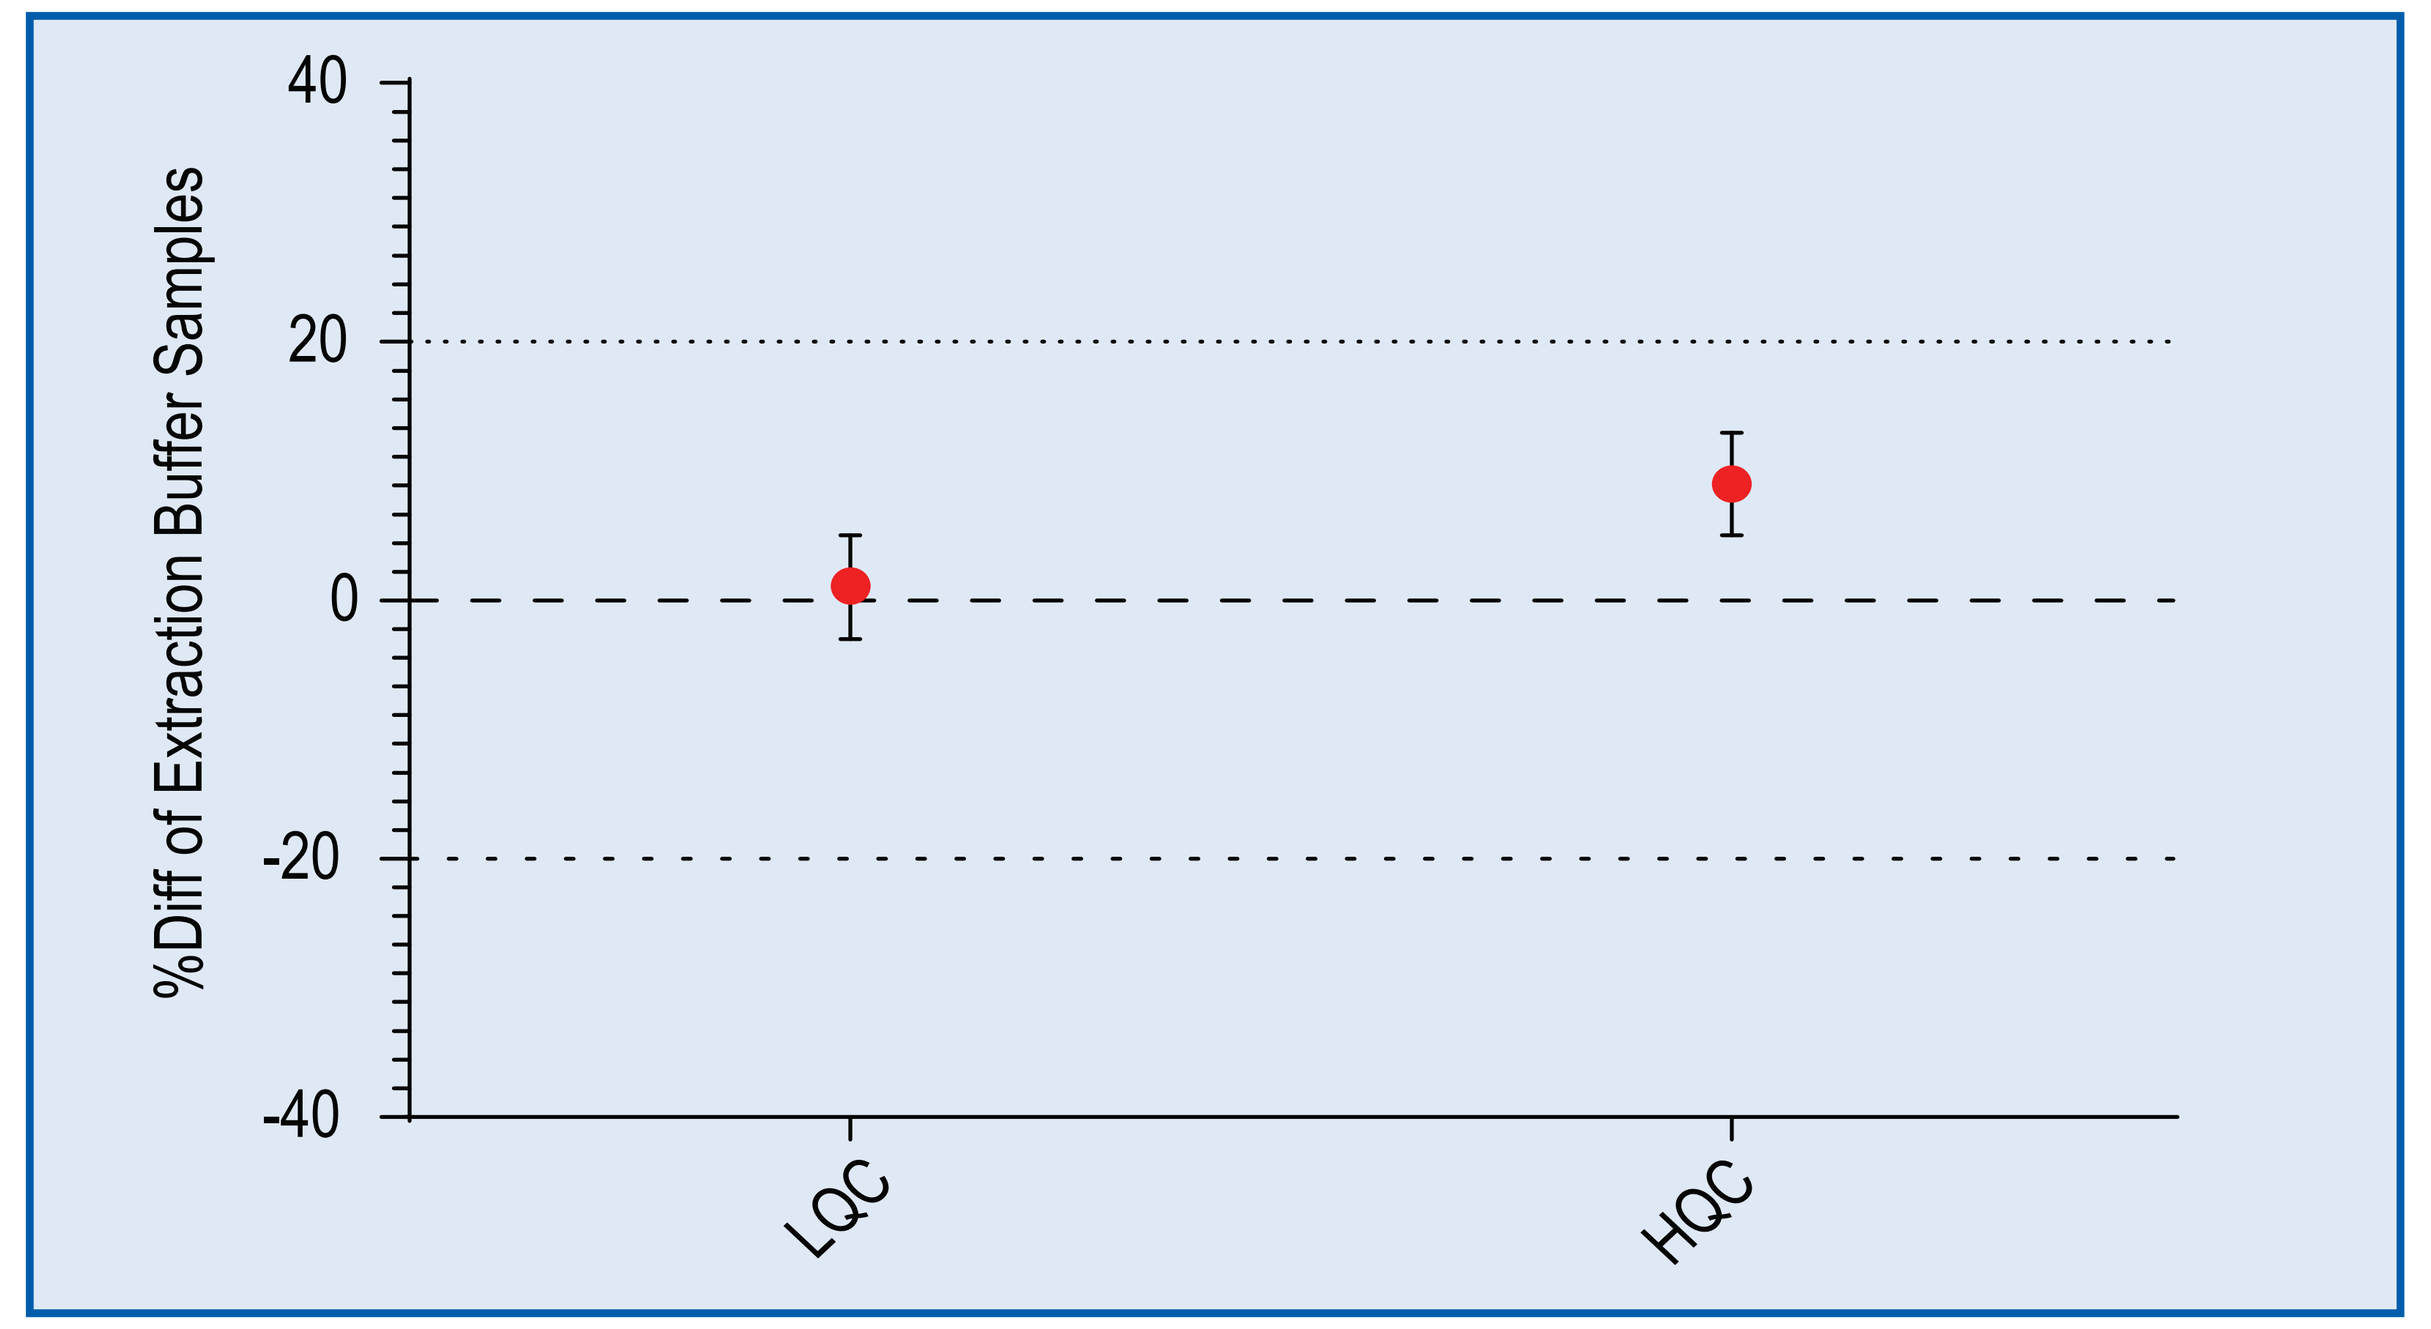

%Diff of Extraction Buffer Samples
40
20
0
-20
-40
LQC
HQC

No blank WB matrix (Sample Diluent) effect was observed

Accuracy and Precision

Standard Data

4 assays, conducted by 2 analysts using varying incubation times, were performed over a 7 day period. All standard curves are superimposed as shown in the Figure 2 below. Accuracy and precision results are listed in Table 2.

Figure 2. Standard Curve Performance

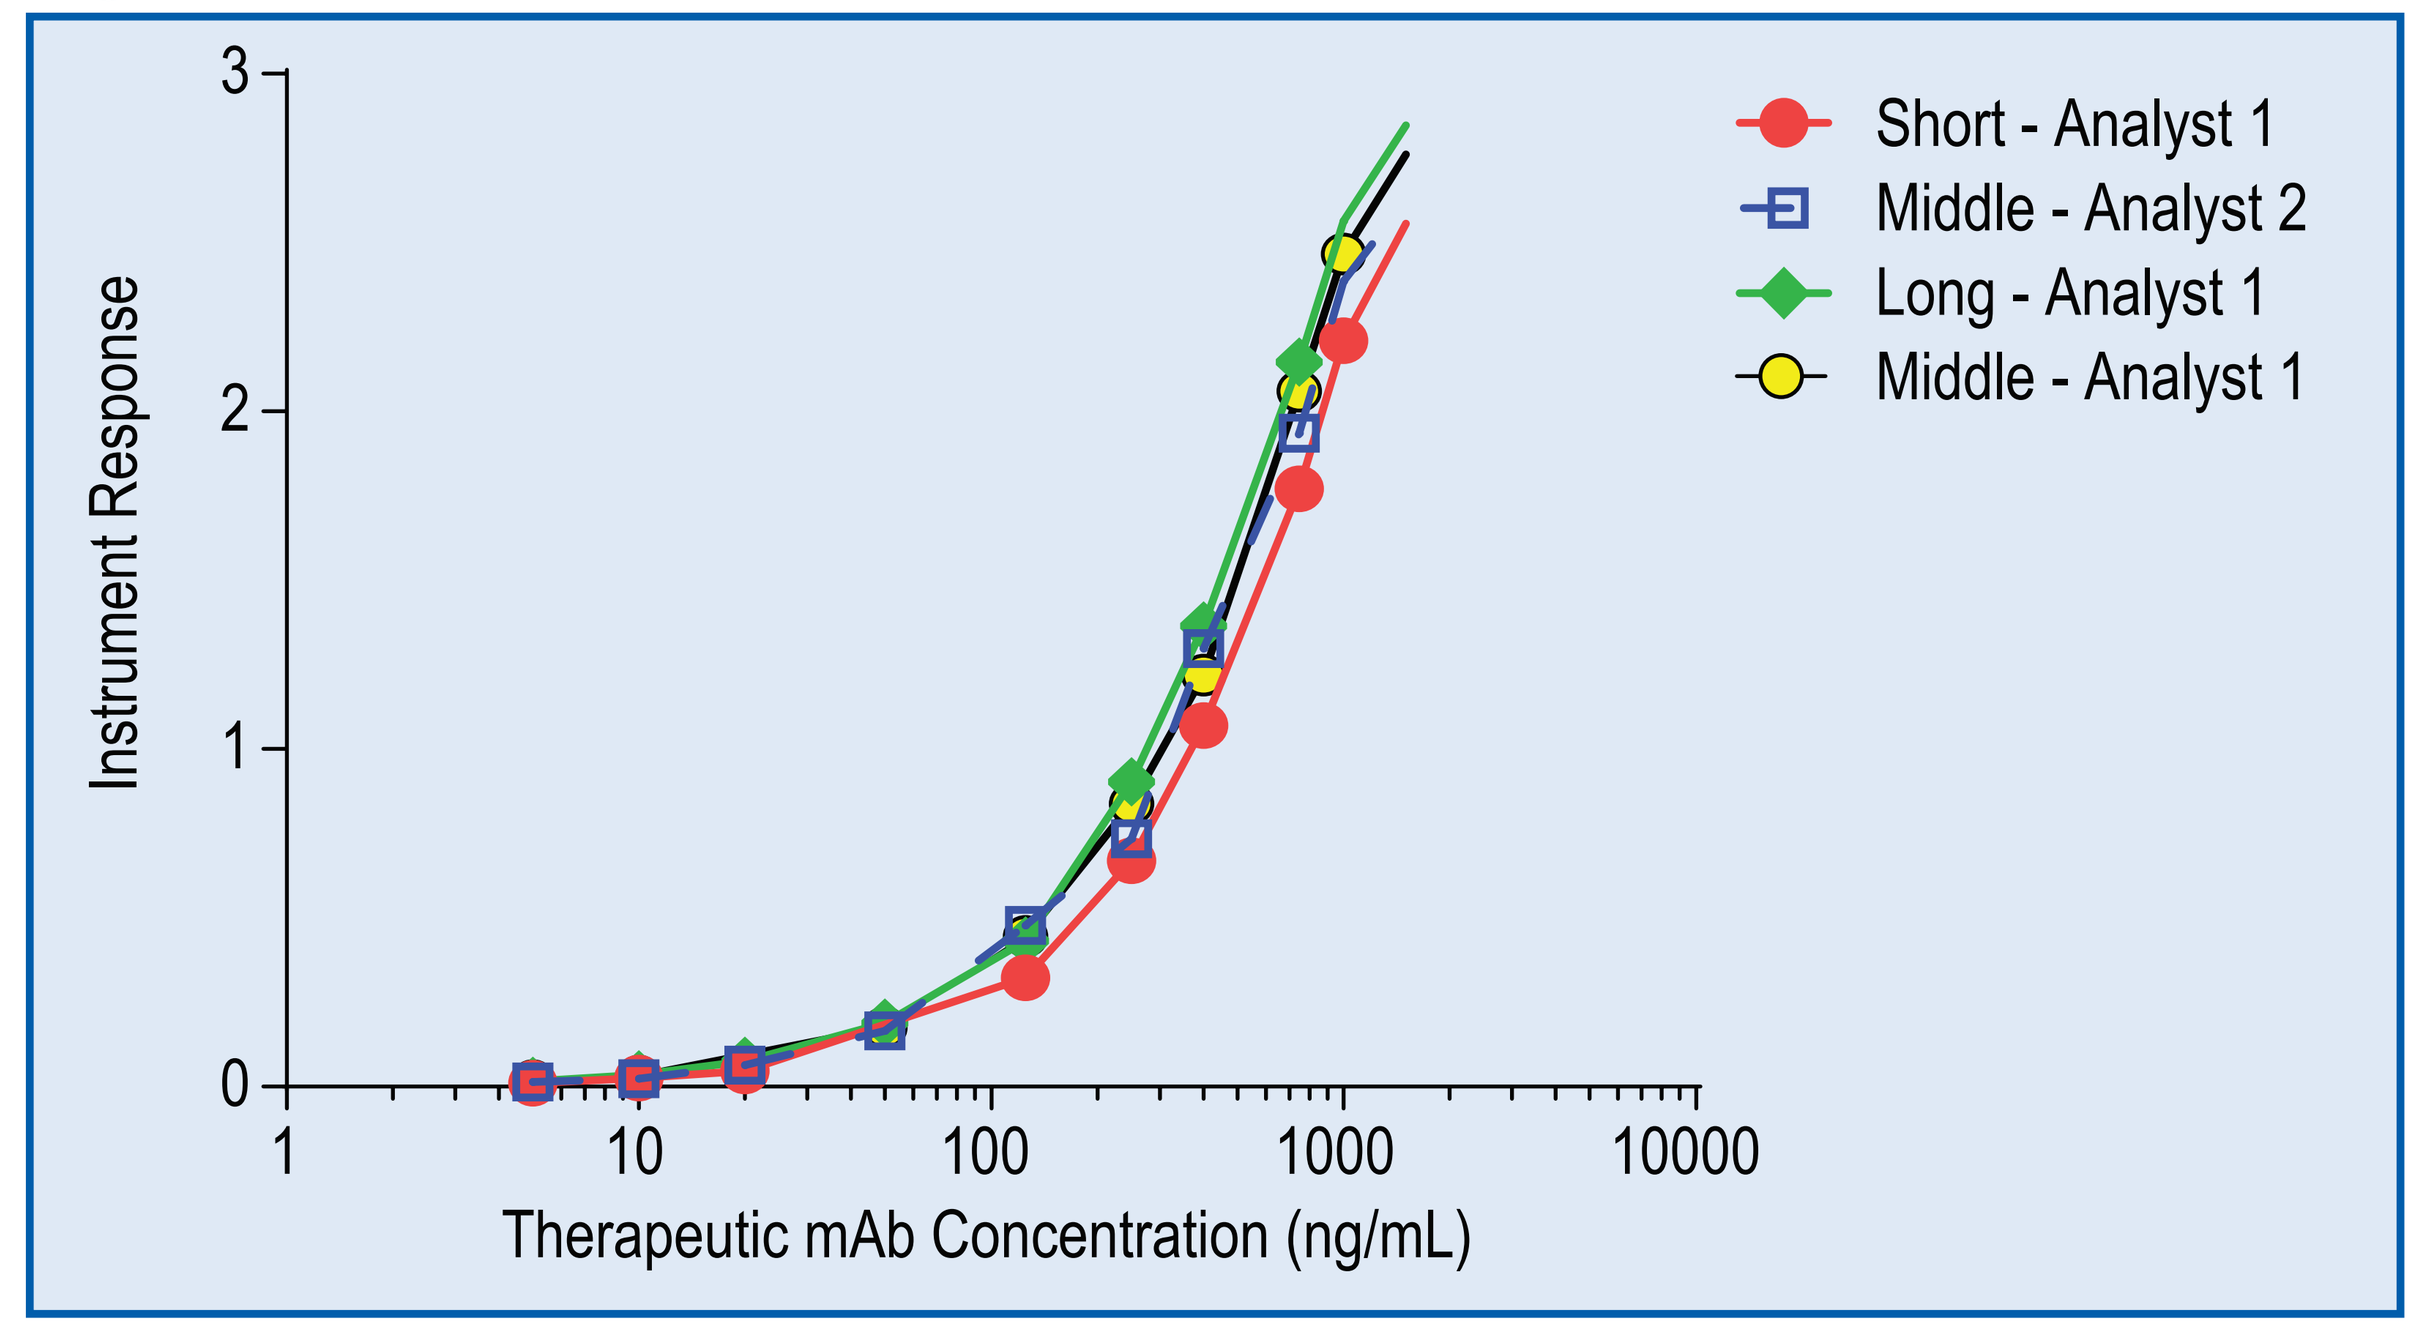

Instrument Response
Therapeutic mAb Concentration (ng/mL)
1
10
100
1000
10000
Short - Analyst 1
Middle - Analyst 2
Long - Analyst 1
Middle - Analyst 1

Table 2. Standard and QC Inter-Assay Data Summary

Sample Type	Accuracy (%Bias)	Precision (%CV)	Total Error (%)
Standards	12.0 to –4.7	12.6 to 1.2	N/A
Quality Controls	7.3 to –7. 2	11.1 to 9.8	18.4 to 10.5

Table 3. Accuracy and Precision Comparison of DBS, VAMS, and Serum Samples

Sample Type	Nominal Concentration (ng/mL)		Inter-assay Accuracy (%RE)			Inter-assay Precision (%CV)			Total Error of Method (%TE)		
	DBS/VAMS	Serum	DBS	VAMS	Serum	DBS	VAMS	Serum	DBS	VAMS	Serum
Standards	10–1000	20–2000	–3.0 to 7.0	–4.7 to 12.0	–3.4 to 8.0	3.8 to 7.0	1.2 to 12.6	0.8 to 3.2	N/A	N/A	N/A
LLQC	10	20	–5.2	7.3	2.6	19.1	11.1	7.1	24.3	18.4	9.8
LQC	30	60	–2.7	5.3	1.9	11.5	10.4	6.9	14.2	15.7	8.8
MQC	200	400	–9.1	–7.2	1.2	7.8	10.2	4.9	16.9	17.4	6.0
HQC	600	1200	–5.1	–0.6	3.4	7.9	9.9	11.9	12.9	10.5	15.3
ULQC	1000	2000	–2.3	4.5	–3.9	9.1	9.8	8.5	11.3	14.3	12.3

VAMS™ samples demonstrated comparable results to DBS samples with an improved total error at LLQC

Selectivity

Samples of WB from 14 different individuals were treated either by not spiking, spiking at LQC, or spiking at LLOQ prior to preparing VAMS™. The VAMS™ were then prepared, extracted, and analyzed. No matrix effect was observed which was consistent with DBS results. The results are listed in table 4.

Table 4. Selectivity Results

Patient Type	Number of Lots Tested	Blank Sample Concentration	% of Lots that Met Method Acceptance Criteria[2]
Normal Males	10	All BQL[1]	100
Normal Females	4	All BQL	100

[1]Below Quantitation Level
[2]Acceptance criteria is at least 80% of tested lots should be within 20% of the spiked nominal concentration

Dilutional Linearity

A WB sample was prepared at expected C_{max} (15 µg/mL) and was diluted using WB Extracted Blank (Sample Diluent) into assay range at 5 different concentrations ranging from 1:15 to 1:1500. The VAMS™ were then prepared, extracted, and analyzed. The dilutional linearity was demonstrated and results were comparable to the DBS evaluation results (see Figure 3 below).

Figure 3. Dilutional Linearity Performance of VAMS™ versus DBS

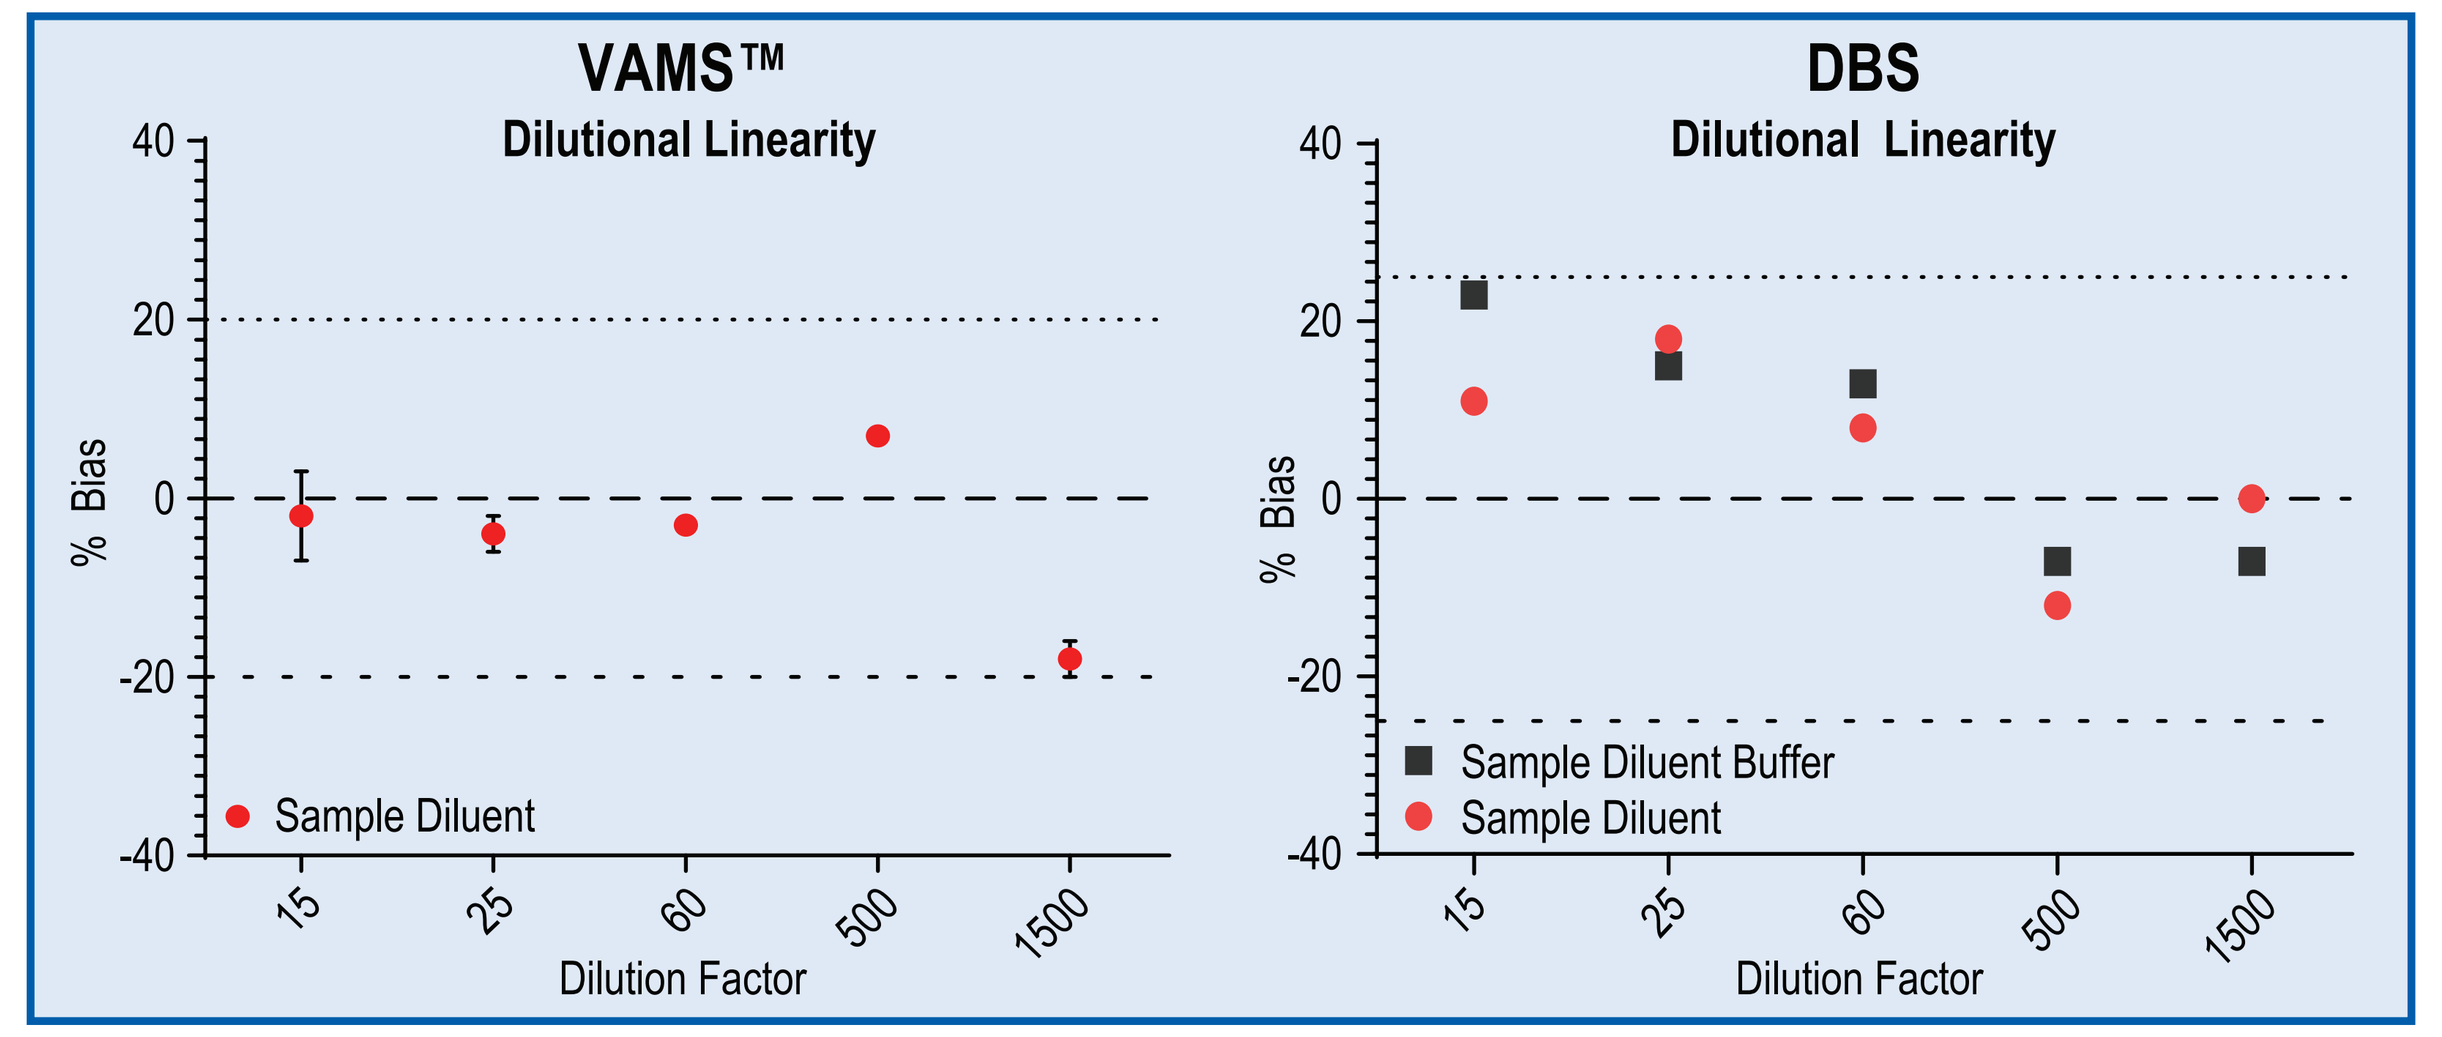

VAMS™
Dilutional Linearity
DBS
Dilutional Linearity
% Bias
Sample Diluent
Sample Diluent Buffer
Sample Diluent
15
25
60
500
1500
Dilution Factor

Hematocrit Effect

Whole blood samples with hematocrit levels of 20% and 60% and a baseline sample with hematocrit of 43% were used for the evaluation. The blood samples were spiked with the therapeutic mAb at HQC and LQC levels. Prepared blood samples were then used for the VAMS™ sample preparation, extraction, and analysis. No hematocrit effect was observed which demonstrated the superb analytical accuracy of VAMS™ technology over DBS technology (see Figure 4).

Figure 4. Hematocrit Effect of VAMS™ versus DBS

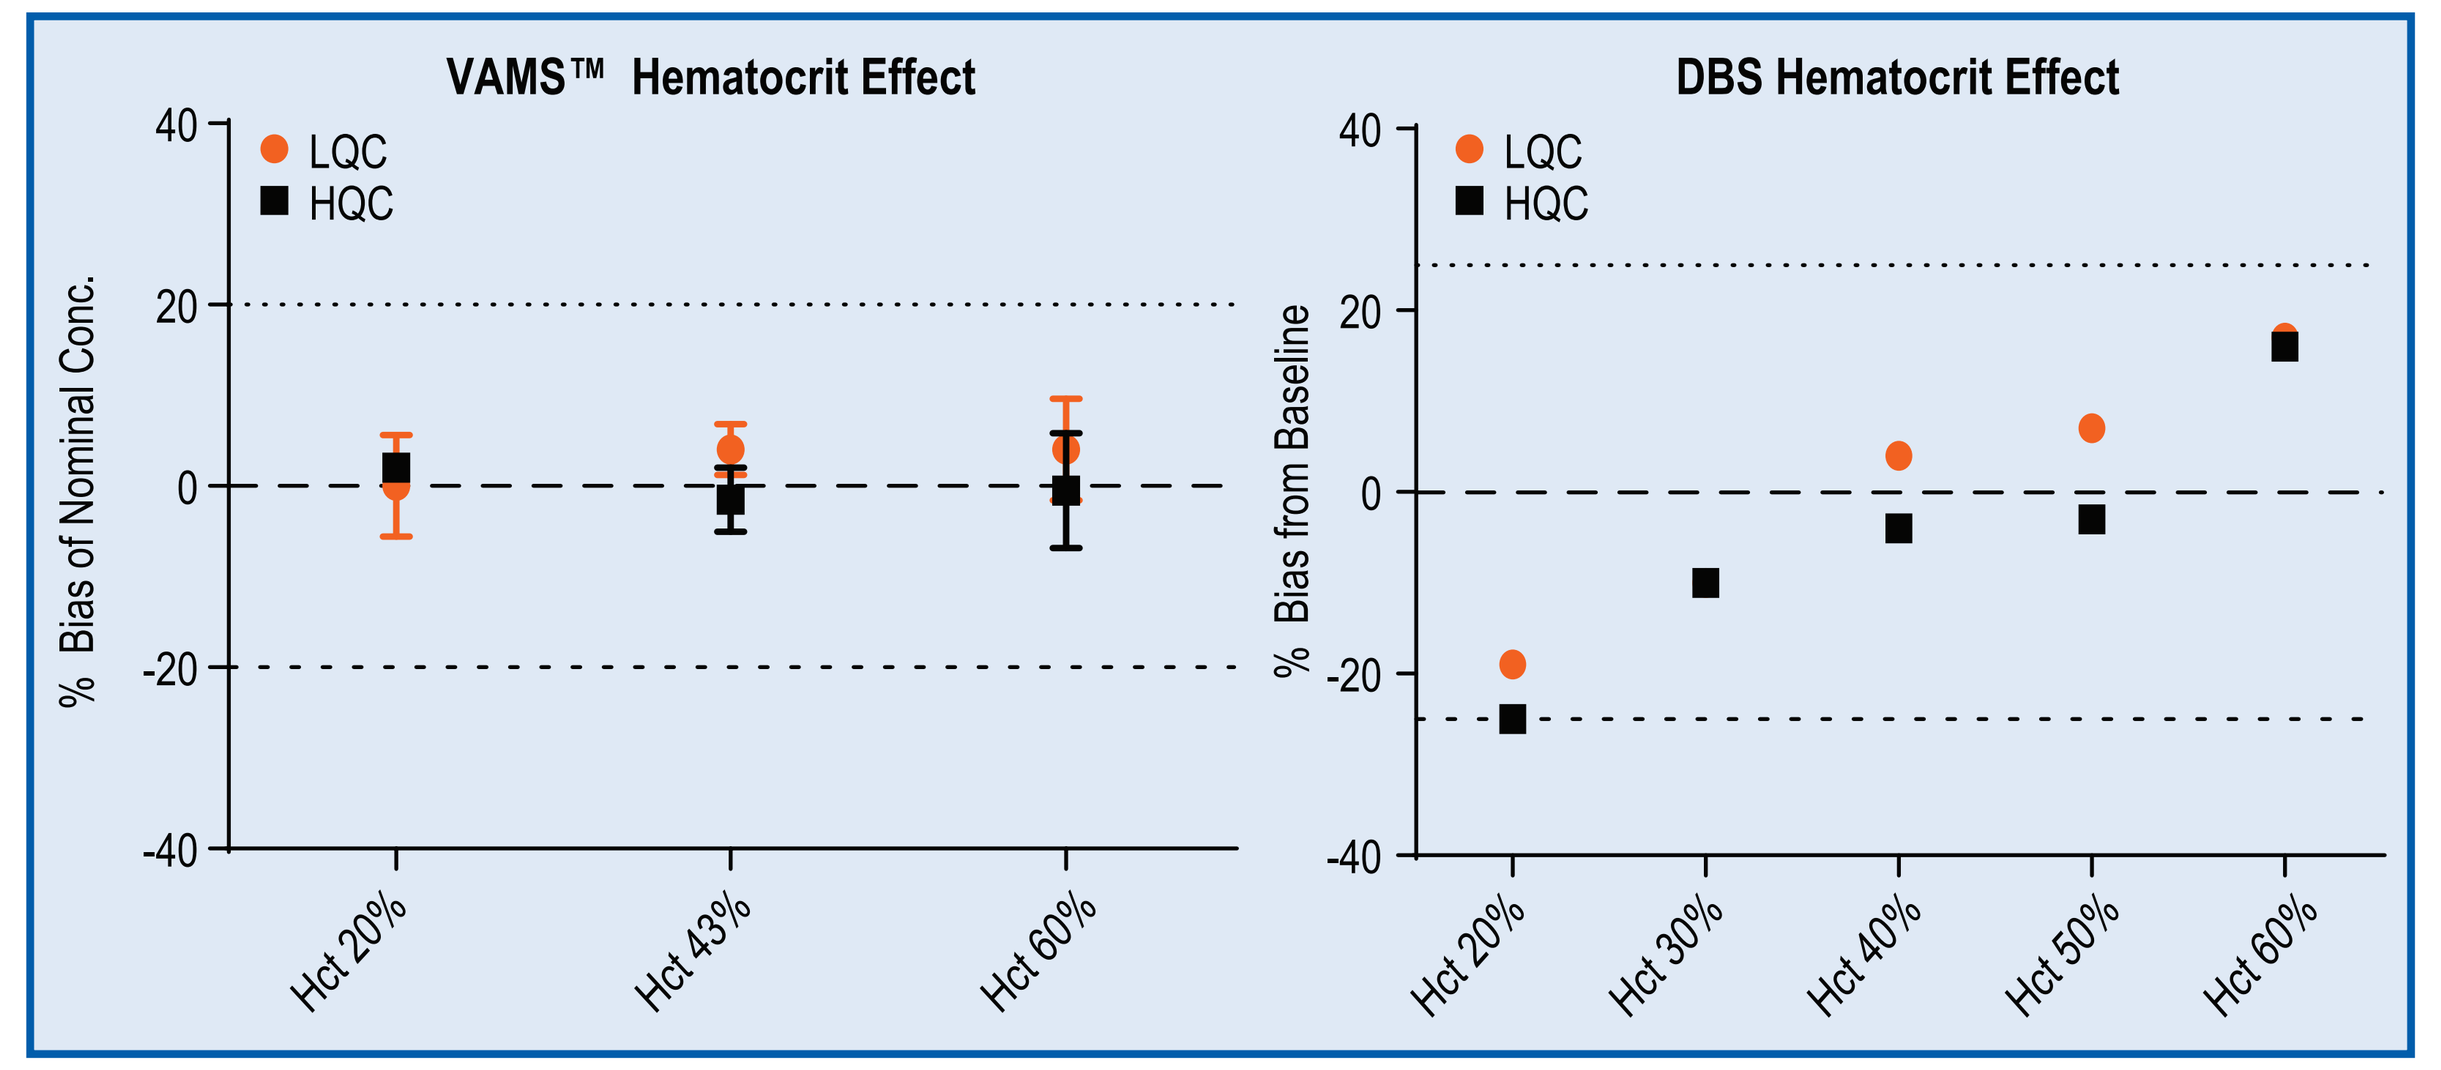

VAMS™ Hematocrit Effect
DBS Hematocrit Effect
% Bias of Nominal Conc.
% Bias from Baseline
LQC
HQC
Hct 20%
Hct 30%
Hct 40%
Hct 43%
Hct 50%
Hct 60%

Long Term Stability

Limited sample bench top process stability and long term sample storage stability evaluations were conducted. Stability (up to 33 days) was demonstrated at all conditions tested (see Figure 5).

Figure 5. Long Term Stability

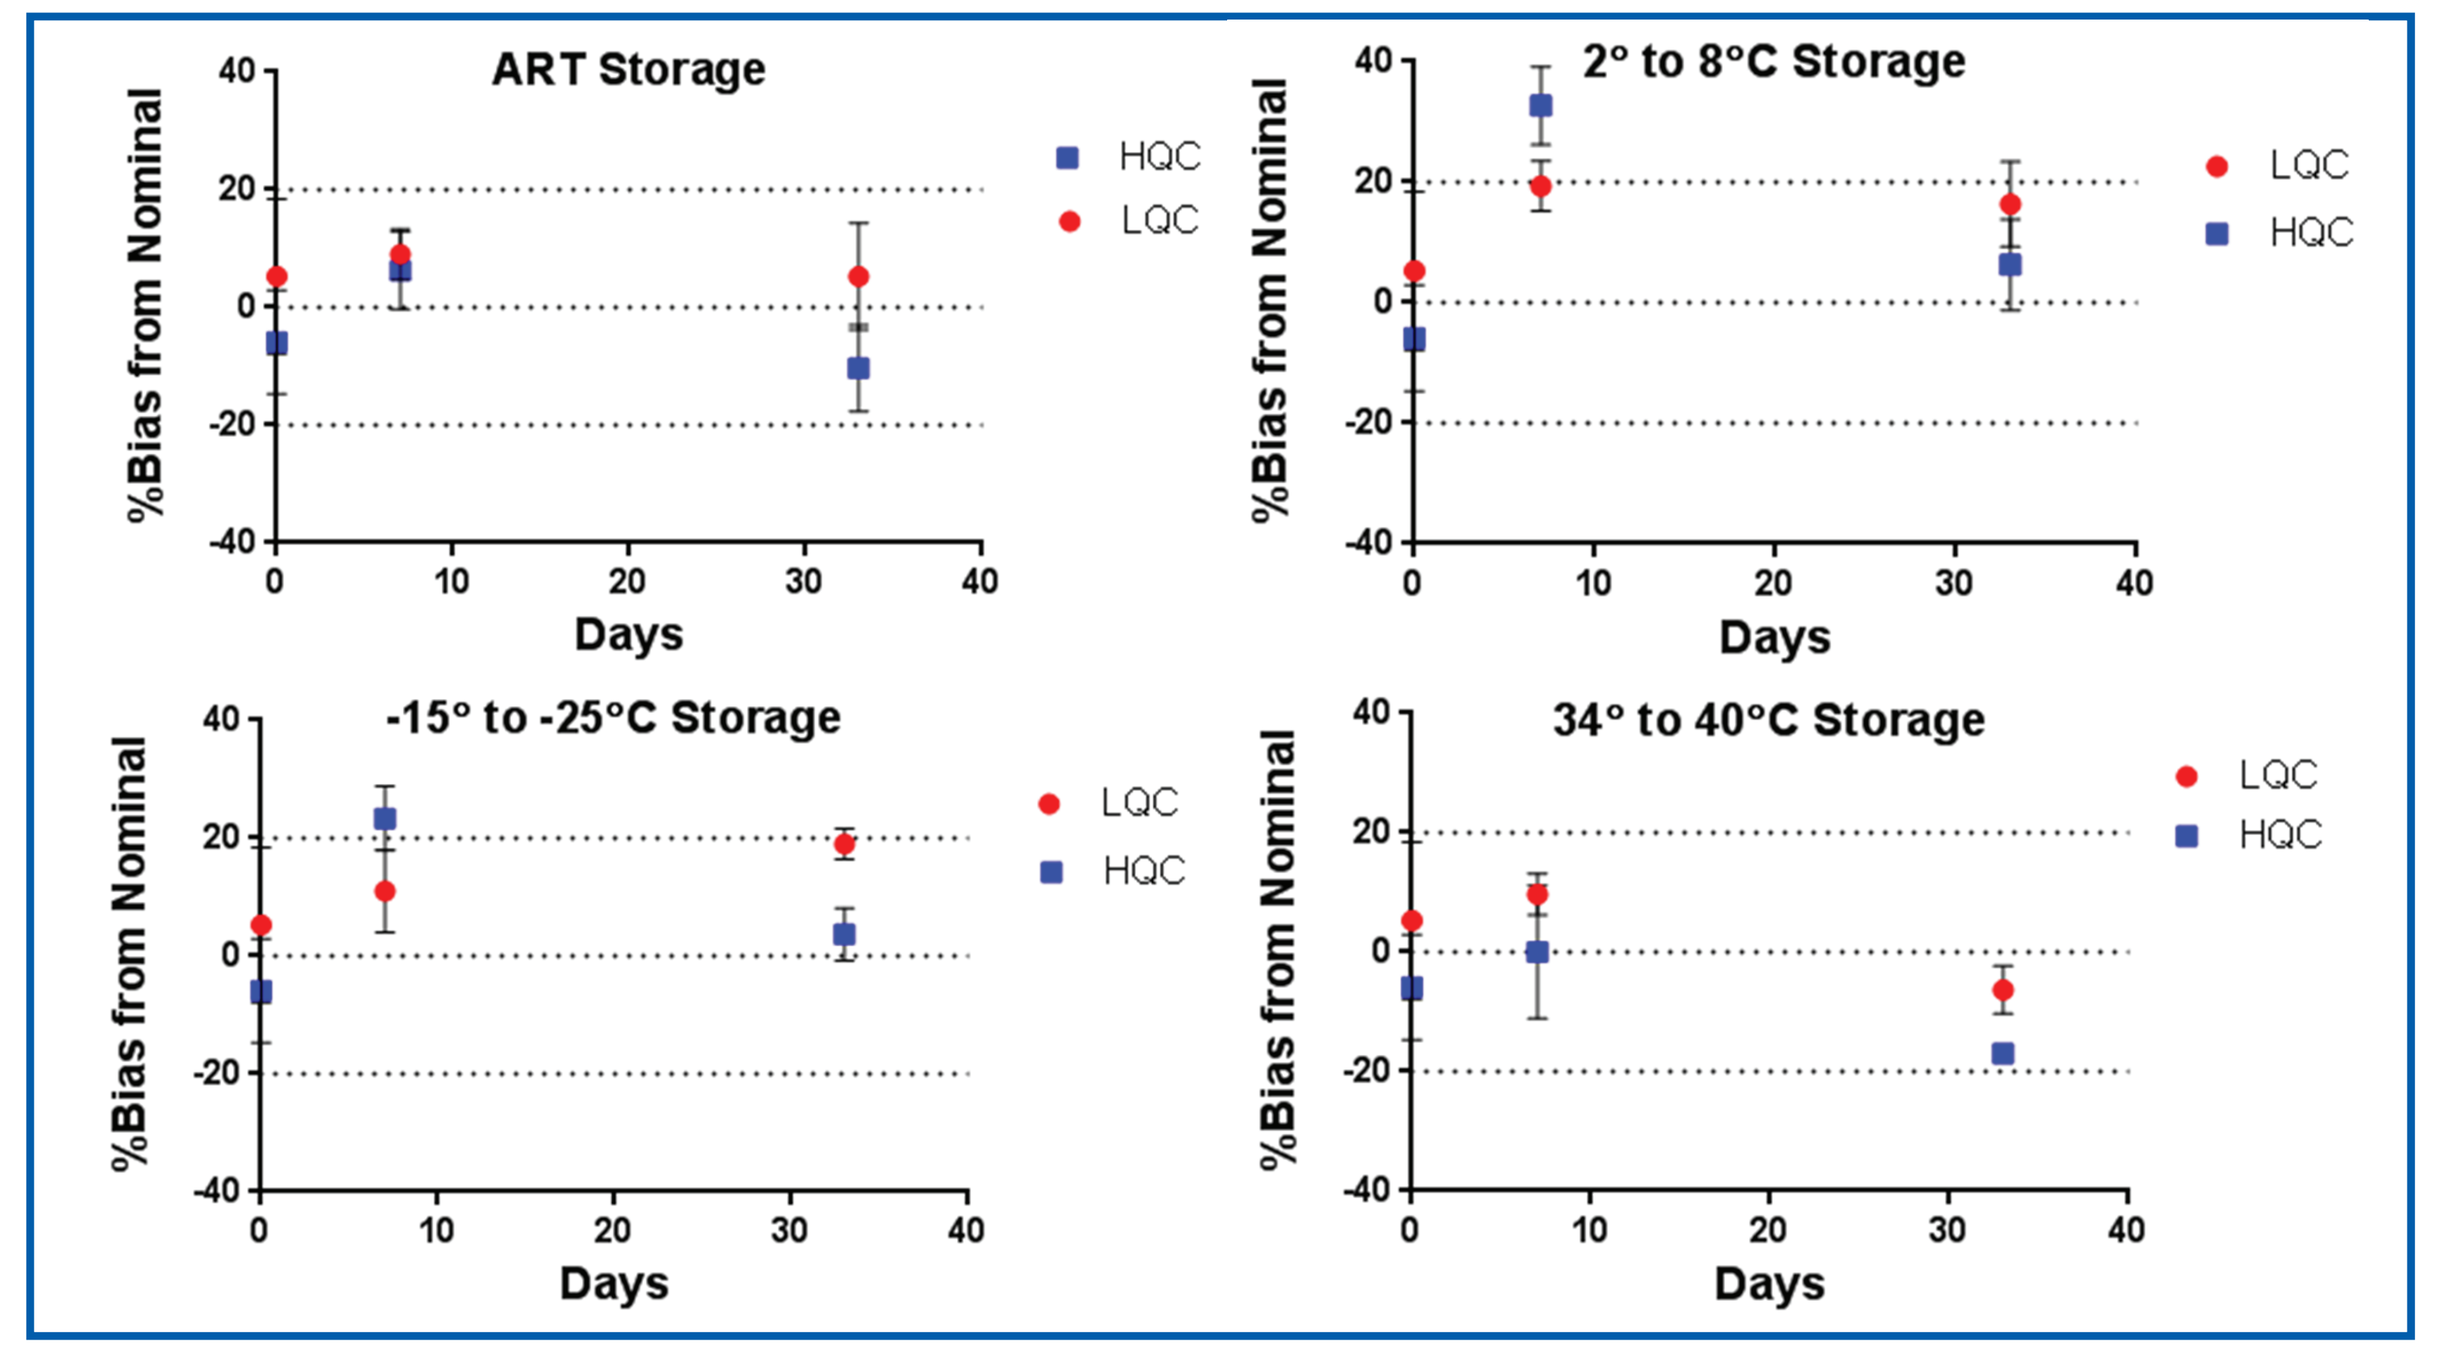

ART Storage
2° to 8°C Storage
-15° to -25°C Storage
34° to 40°C Storage
%Bias from Nominal
Days
LQC
HQC

CONCLUSIONS

- We successfully evaluated the VAMS™ technology using a Ligand Binding Assay. The analytical results generated with the use of VAMS™ were comparable to the results of using DBS technology. The VAMS™ technology provided additional advantages in simplified sample processing and better performance in certain validation parameters:
 - VAMS™ technology did not require labor-intensive pipetting and coring steps as did the DBS technology.
 - VAMS™ sampling technique did not demonstrate hematocrit effect while hematocrit effect was a major concern for DBS technology.
 - VAMS™ technology demonstrated improved accuracy and precision results with a better total error at LLOQ.
- Overall, based on this preliminary evaluation, VAMS™ technology is a viable alternative to DBS with some added improvements.

ACKNOWLEDGEMENT

We would like to acknowledge Phenomenex® for developing this technology and providing the materials necessary to perform this evaluation.

REFERENCES

Daniel Burns, Laura Brunner, Surendran Rajendran, Beth Johnson, Mark Ma, and Jin Wang. Validation of a Ligand Binding Assay Using Dried Blood Spot Sampling. *The AAPS Journal*, Vol. 15, Issue 1. 123-131 (2013)

AMGEN © Amgen Inc. 2015

AAPS National Biotechnology Conference, San Francisco, CA; June 8–10, 2015