DEVELOPMENT AND VALIDATION OF A VOLUMETRIC ABSORPTIVE MICROSAMPLING (VAMS) APPROACH FOR DETERMINATION OF A NOVEL ANTIEPILEPTIC DRUG IN HUMAN DRIED BLOOD

S.Wood[1], K.Holmes[1], I.Morelli[1], J.Segelbacher[2] and C.Rospo[3]

1 Unilabs York Bioanalytical Solutions, Northminster Business Park, York, YO26 6QR, UK

2 Unilabs York Bioanalytical Solutions, Discovery Park, Ramsgate Road, Sandwich, Kent, CT13 9ND, UK

3 UCB Biopharma S.P.R.L, Chemin du Foriest B-1420 Braine-l'Alleud, Belgium

Introduction

There is increasing interest within clinical drug development for using microsampling techniques to allow small volume sample collection. The potential advantages of dried blood microsampling have been well documented and include applicability to compliant home sampling and application to paediatric sampling, cost effective sample shipment and storage. Volumetric absorptive microsampling (VAMS™) provides a potentially viable option for obtaining such dried blood samples without the known issues with more conventional paper based techniques.
Methods to support the determination of the drugs in dried blood obtained using VAMS need to be individually developed and validated and have their own unique challenges compared to more traditional wet plasma assays. Here we describe the development and validation of an LC-MS/MS method to determine a novel antiepileptic drug in human dried blood to support a clinical study aimed at bridging between dried blood microsampling and wet plasma sampling. The VAMs microsampling utilised Mitra® devices to obtain fixed volume dried blood samples.
This study was funded by UCB BIOPHARMA SPRL, Belgium.

Method Development

Prior to validation, method development was performed to determine the most suitable conditions for the extraction of the analyte from the Mitra® tips.
Analyte working solution was spiked into fresh blood, mixed and immediately sampled onto multiple tips. Tips were then dried in the rack overnight.

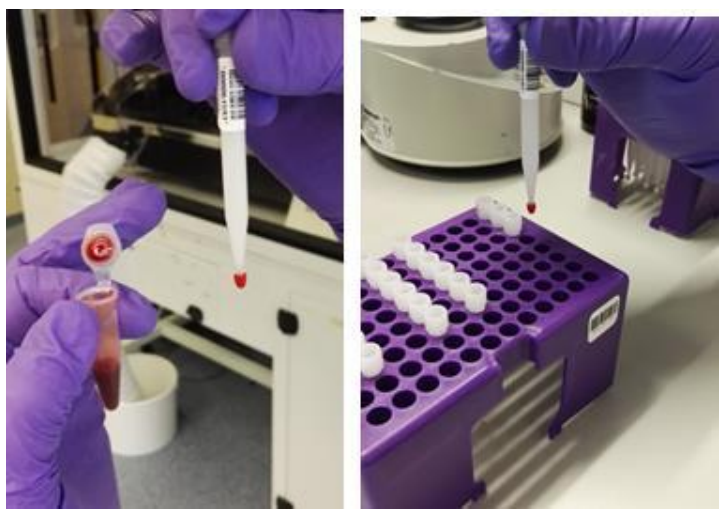

Figure 1: Sampling of blood onto the tips and into the 96 rack for drying.

Recovery solutions were prepared for comparison based on a 10 µL sample on the tip. The two approaches that were tested initially are summarised in table 1.

Dried Tips Submerged for Extraction	
1: Organic	**2: Aqueous**
300 µL Methanol (+acid)	150 µL Water (+acid)
Shake and remove tips	Shake and remove tips
-	Protein precipitation (add 500 µL acetonitrile)
Centrifuge and transfer	Centrifuge and transfer
(dry down/reconstitute)	(dry down/reconstitute)
Inject onto LC-MS/MS	Inject onto LC-MS/MS
45% recovery	100% recovery

Table 1: Two alternate approaches for extracting the analyte from blood dried on tips.

Option two showed the better recovery and was selected for further optimisation.
Different protein precipitation crash solvents all gave good recovery, although lower than observed in plasma.

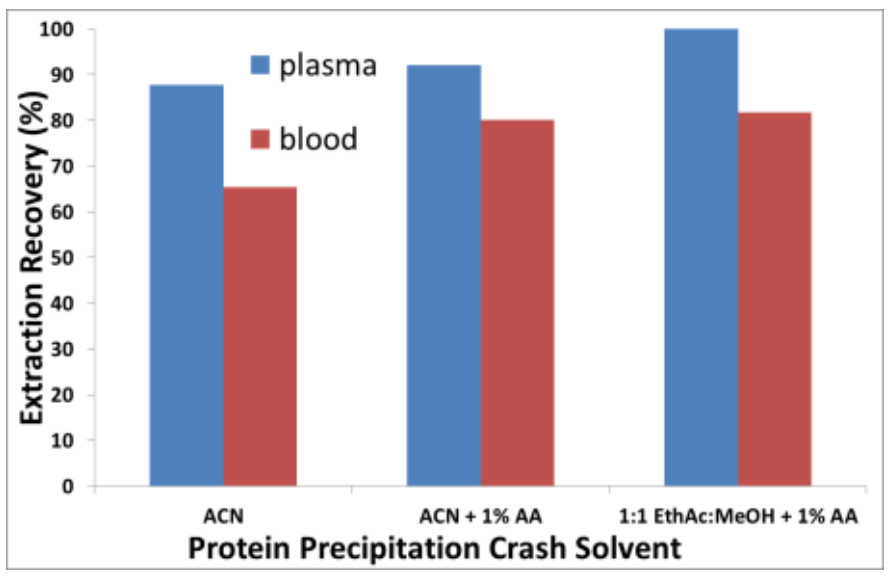
plasma
blood
Extraction Recovery (%)
100
90
80
70
60
50
40
30
20
10
0
ACN
ACN + 1% AA
1:1 EthAc:MeOH + 1% AA
Protein Precipitation Crash Solvent

Figure 2: Comparison of crash solvents (acetonitrile (ACN) or 1:1 ethyl acetate:methanol (EthAc:MeOH) and acetic acid (AA)) for protein precipitation from plasma and dried blood on Mitra tips extracted into water.

In addition, the following were investigated for the impact on recovery:

- Amount of time shaking in water for extraction from the tips – 20, 40 and 60 minutes were tested and gave equivalent results (on tips dried overnight)
- Impact of warming blood prior to sampling – no improvement was made by including this step

During stability testing, lower recovery was observed from tips that had increased drying / storage times, steps were included to improve the extraction:

- Addition of acid (0.1% formic acid) to the water
- Sonication step included with the tips submerged in the water + 0.1% formic acid
- The shaking time that followed was increased to 60 minutes

Effect of Blood haematocrit

Effect of blood haematocrit on quantification of analyte was assessed at the medium QC level. QCs were prepared in fresh control blood with varying haematocrit (30%, 50% and 70%), six replicates per haematocrit level were analysed and compared to nominal concentration. The precision and bias were within the validation acceptance criteria at all haematocrit levels. Although all data were acceptable, a slight trend was observed, thought to be due to decreased extraction recovery at higher blood haematocrit levels.

Nominal conc. (ng/mL)	Haematocrit (%)	Measured mean conc. (ng/mL)	Precision (CV%)	Bias (%)
150	30	158	4.7	5.3
	50	151	4.6	0.7
	70	145	3.8	-3.3

Table 2: Haematocrit bias assessment

Assay Performance

The method was fully validated over the range 2-2000 ng/mL using LC-MS/MS detection.

	Analyte QC nominal concentration (ng/mL)				
	2.00	6.00	150	1600	2000
	Mean intra-run statistics				
Overall intra-run mean (ng/mL)	1.94	6.22	158	1710	1940
Overall intra-run CV (%)	7.2	6.4	5.2	5.4	2.9
Overall intra-run bias (%)	-3.0	3.6	5.5	6.9	-3.2
	Inter-run statistics				
Inter-run mean (ng/mL)	1.94	6.22	158	1710	1940
Inter-run SD (n-1)	0.174	0.387	10.1	92.2	95.3
Inter-run CV (%)	9.0	6.2	6.4	5.4	4.9
Inter-run bias (%)	-3.0	3.7	5.3	6.9	-3.2
n	18	18	18	18	18

Table 3: Validation data for inter-run and intra-run precision and bias

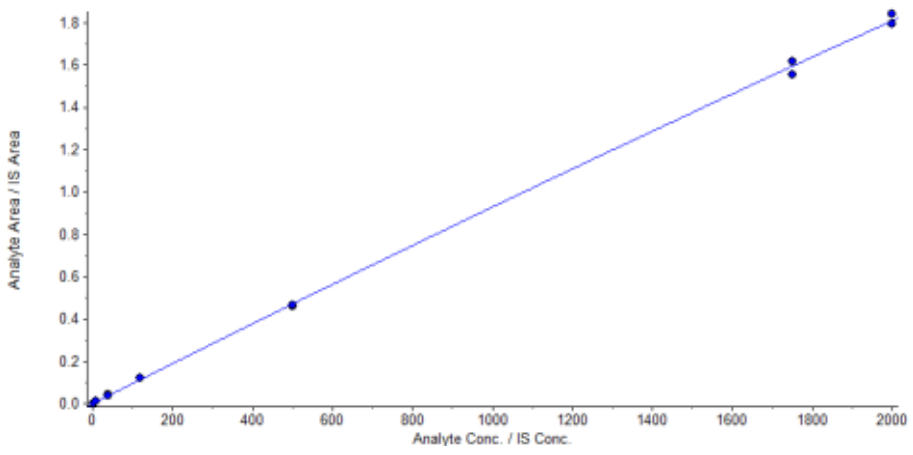
Analyte Area / IS Area
Analyte Conc. / IS Conc.

Figure 3: Assay linearity. Calibration standard curve Range 2.00 - 2000 ng/mL, a quadratic regression with a weighting of 1/x was used

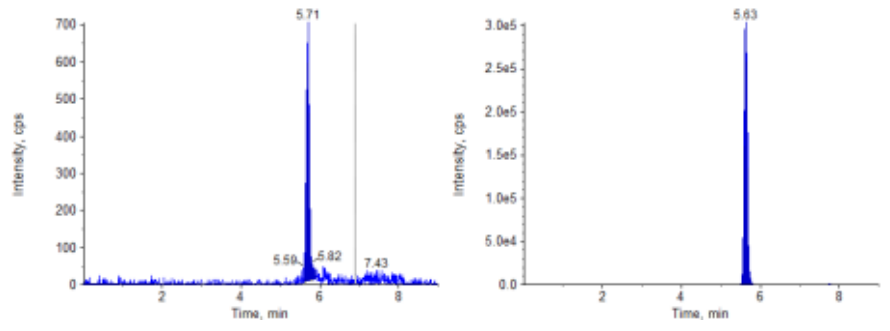
5.71
5.59
5.82
7.43
5.63
Intensity, cps
Time, min

Figure 4: Assay sensitivity. Chromatogram of LLOQ calibration standard (2.00 ng/mL)

Short term storage assessments

In order to assess sample integrity under potential 'ambient' sample shipping, dry blood samples were subjected to storage at elevated (40°C) and lowered (-20°C) temperatures for 48 hours. Stability was demonstrated at both storage conditions for Low, High and Dilution QC levels

Nominal Analyte concentration (ng/mL)	Samples stored for 48 hours at 40°C: Mean Observed concentration (ng/mL)	Difference from nominal (%)
6.00	6.09	1.5
1600	1840	15.0
5000	5020	0.4

Table 4: QC stability at elevated temperature

Nominal Analyte concentration (ng/mL)	Samples stored for 48 hours at -20°C: Mean Observed concentration (ng/mL)	Difference from nominal (%)
6.00	6.22	3.7
1600	1720	7.5
5000	5000	0.0

Table 5: QC stability at lowered temperature

Calibration Standards/QC sample preparation

Calibration standards and QC samples were prepared by spiking analyte into whole blood (LiH), 10 µL Mitra tips were allowed to lightly touch the surface of the blood to completely coat the tip by capillary action. It was found that the tips could be dipped in bulk by preparing and sub aliquotting ~80 µL of blood into each well of a 96-well plate and dipping a full Mitra autorack in one go, this allowed for efficient preparation of validation samples. Mitra tips were supplied uniquely barcoded as standard, providing a means to easily identify and document preparations

Drying time

As part of method validation an investigation was carried out to assess the minimum time to allow for drying of fresh calibration standards before extraction. QC samples which were prepared one week previously were analysed against fresh calibration standards which had been subjected to 3 hour and 24 hour drying times. The data showed QC's analysed against calibration standards left to dry for 3 hours prior to analysis had an unacceptable negative bias. When compared to standards left to dry for 24 hours, the bias was acceptable. This is thought to be due to increased recovery of the calibration standards where the 3 hour drying time was deemed to be insufficient. A minimum 'overnight' drying time was implemented and was sufficient for all subsequent analysis.

Calibration standard drying time	Nominal QC concentration (ng/mL)					
	6.00	%bias	150	%bias	1600	%bias
3 hours	3.95	-34.2	117	-22.0	1358	-15.1
24 hours	6.25	4.2	156	4.0	1550	-3.1

Table 6: Effect of sample drying time

Conclusions

Method optimisation for VAMS based methods has special considerations associated with obtaining consistent recovery from dried samples with minimal variation with respect to blood haematocrit and dried sample age.
Method validation needs to take into account additional sample variables associated with sample shipment conditions and sample age.
The validated method was applied to human dried blood samples derived from a single and multiple dose study aimed at bridging VAMS to plasma sampling. The method performed with a 100% success rate and generated high quality pharmacokinetic data.

Inspired by **patients.** Driven by **science.**

Unilabs York Bioanalytical Solutions